BAER-101, a selective potentiator of α2- and α3-containing $GABA_A$ receptors, fully suppresses spontaneous cortical spike-wave discharges in Genetic Absence Epilepsy Rats from Strasbourg (GAERS)

Amy S. Chappell[1], Alexis Evrard[2], Hugo Monchal[2], Corinne Roucard[2], and Alexandra MacLean[1],
[1]Avenue Therapeutics, Miami, FL, USA; [2]SynapCell SAS, Saint Ismier, France

Introduction

BAER-101 (formerly AZD7325, see chemical structure below) is a selective partial potentiator of α2/3-containing $GABA_A$ Receptors (GABAARs) and produces minimal sedation and dizziness. Antiseizure effects in models of Dravet and Fragile X Syndromes have been published. BAER-101 has been administered to over 700 healthy human volunteers and patients where it was found to be safe and well tolerated. To test the extent of the anti-seizure activity of BAER-1010, we used the widely-used and translationally relevant Genetic Absence Epilepsy Rats from Strasbourg (GAERS) model. This rat strain exhibits spontaneous generalized cortical seizures in the form of spike-wave discharges (SWDs) during an absence seizure in which there is behavioral arrest (**Figure 1**).

Structure of BAER 101

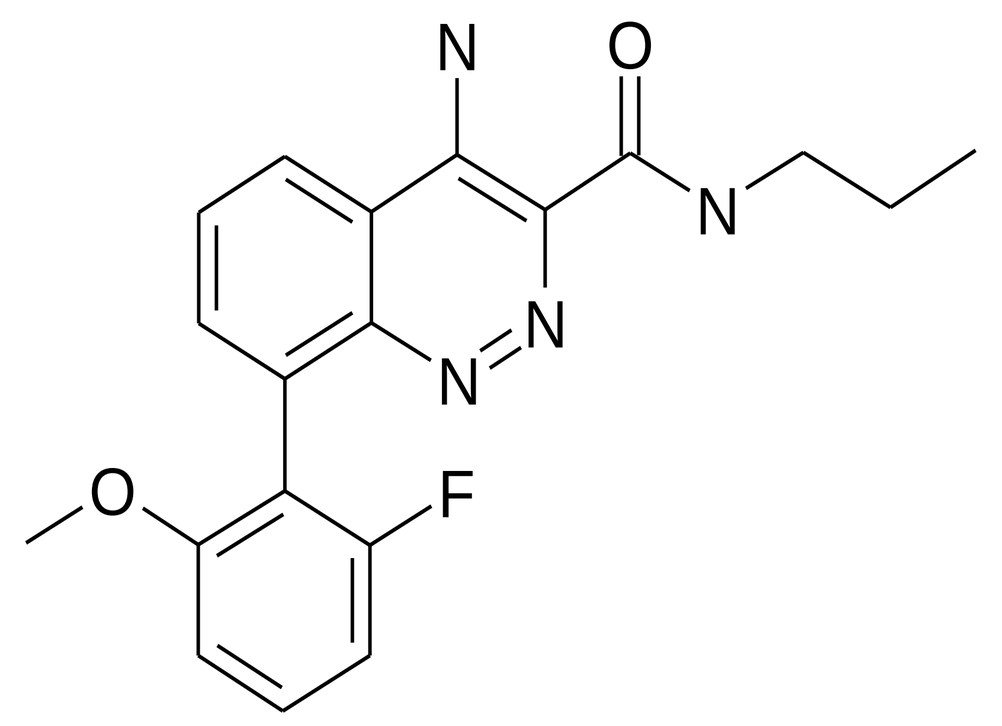

FIGURE 1. A representative EEG tracing showing the well-defined spike-wave discharges that occur spontaneously in the GAERS model during the generalized absence seizure period in these rats.

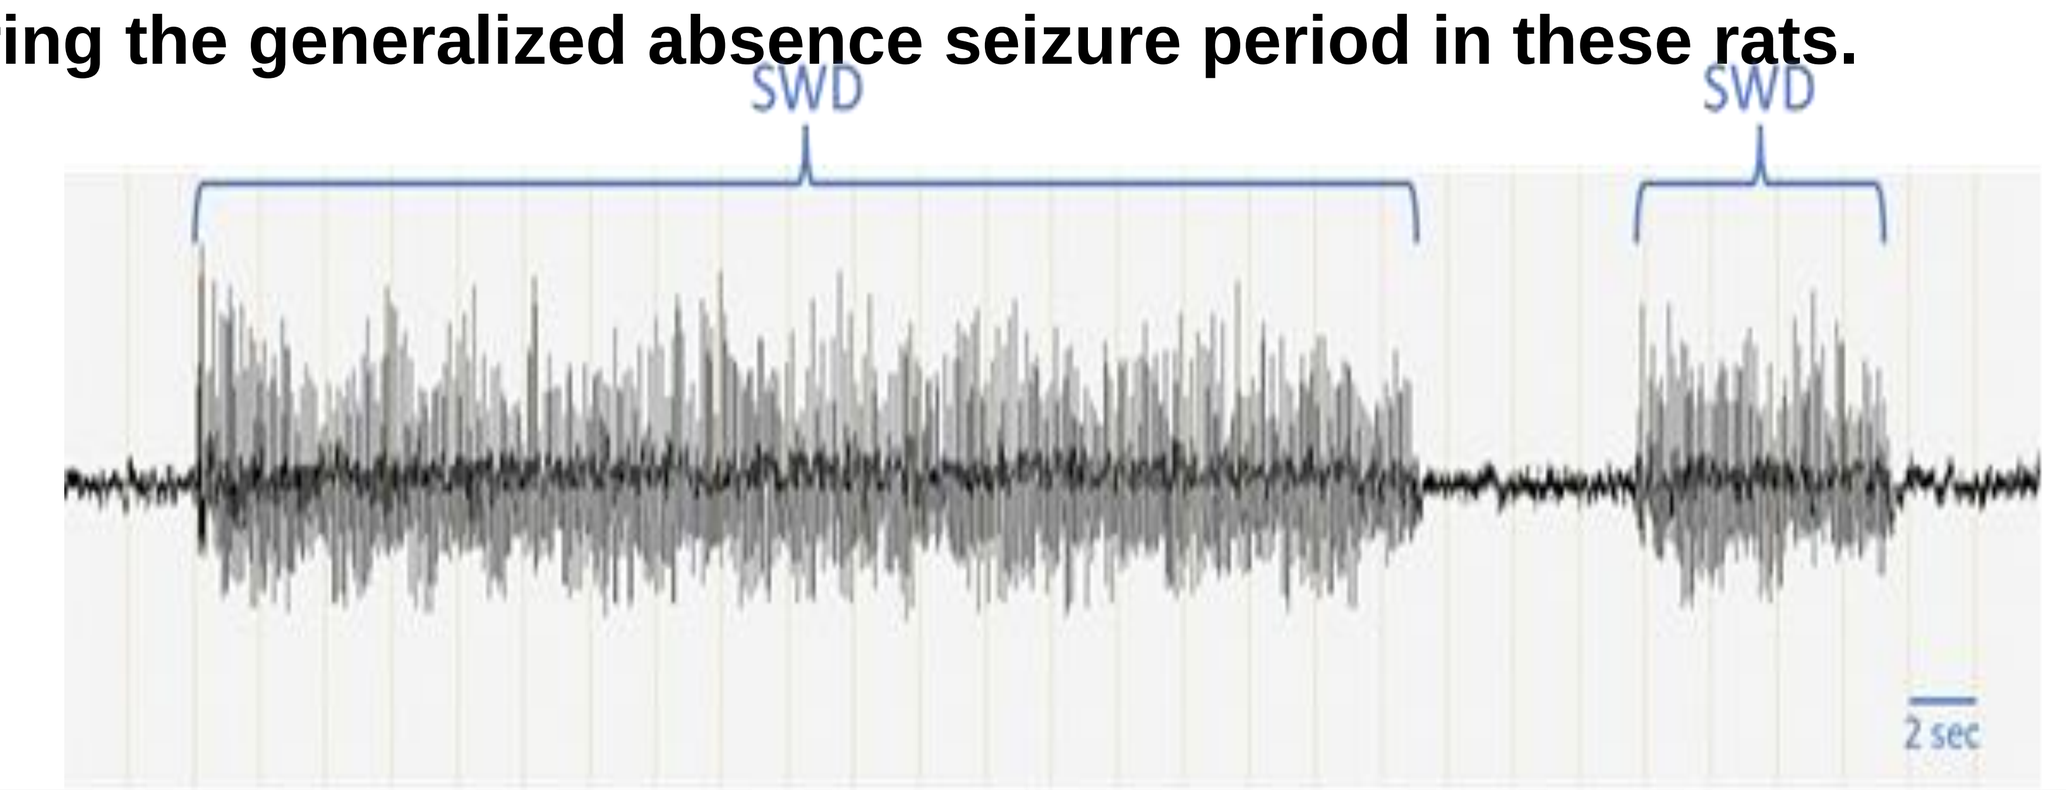
SWD
SWD
2 sec

Study Design

14 experimentally naïve, adult male GAERS (age 4 months) were obtained from Dr Antoine Depaulis (INSERM, Grenoble Institute of Neurosciences, Grenoble, France). Electrodes for spike-wave discharges (SWD) recording were implanted in all 14 rats, under general anesthesia (isoflurane; 2% in oxygen) using stereotaxic methods. There were two phases to the study. The initial phase included the following 6 conditions: Vehicle (20% HPbCD in water) PO, diazepam at 2 mg/kg IP, BAER-101 at 100 mg/kg PO, BAER-101 at 30 mg/kg PO, BAER-101 at 10 mg/kg PO, and BAER-101 at 3 mg/kg PO. Compound conditions were administered in a cross-over protocol, each animal received each condition, in a random order and in separate recording sessions, two animals received each condition in each recording session, and a minimal wash-out period of 7 days was allowed between each administration.

The second phase was four EEG recordings completed with the following set of compound conditions: Vehicle (20% HPbCD in water) PO, diazepam 2 mg/kg IP, and BAER-101 (0.1, 0.3, 1, and 3 mg/kg, PO), administered in a cross-over protocol, following the rules: each animal received 4 different conditions from the 6 indicated above, in a random order and in separate recording sessions, animals having received the vehicle, diazepam or BAER-101 at 3 mg/kg during phase 1 were not ascribed again to these conditions, and a minimal wash-out period of 7 days was allowed between each administration. Drug concentrations were determined for the purpose of correlating with pharmacodynamic effects.

Statistical Methods

EEG recordings were analyzed and quantified blindly to identify SWD. Absolute data measured per 20 min periods were analyzed using a two-way ANOVA with the compound condition and the time from administration as factors. Consequently, measures were considered repeated within the time factor. When significant, ANOVA was followed by paired comparisons using a Dunnett test, with comparisons versus the second baseline period, versus vehicle, and versus diazepam. Pooled data over the whole post-treatment period were analyzed using a one-way ANOVA, with the compound condition as factor. When significant, the ANOVA was followed by paired comparisons using a Dunnett test, versus the vehicle condition.

Results

Diazepam significantly reduced the number of SWD's immediately after administration (**Figure 2**). After evaluation of results from the first 2 recordings, a near complete inhibition of SWD was seen with all doses. BAER-101 induced a dose-dependent inhibition of SWDs, and induced a significant reduction of events at all doses above 1 mg/kg PO, as compared to both the baseline and the vehicle. The effect was immediate and significant during the first measurement time-point, 10 to 90 min after administration, and remained stable over the post-treatment period.

Figure 2: Number of spike-wave discharges (SWD) per 20 min, in the vehicle condition (dark blue), diazepam at 2 mg/kg IP (light blue), and BAER-101 at 0.1 to 100 mg/kg PO (other colours). The grey arrow indicates the time of compound administration. #, ##, ###, ####: $p < 0.05$, 0.01, 0.001 and 0.0001 as compared to vehicle

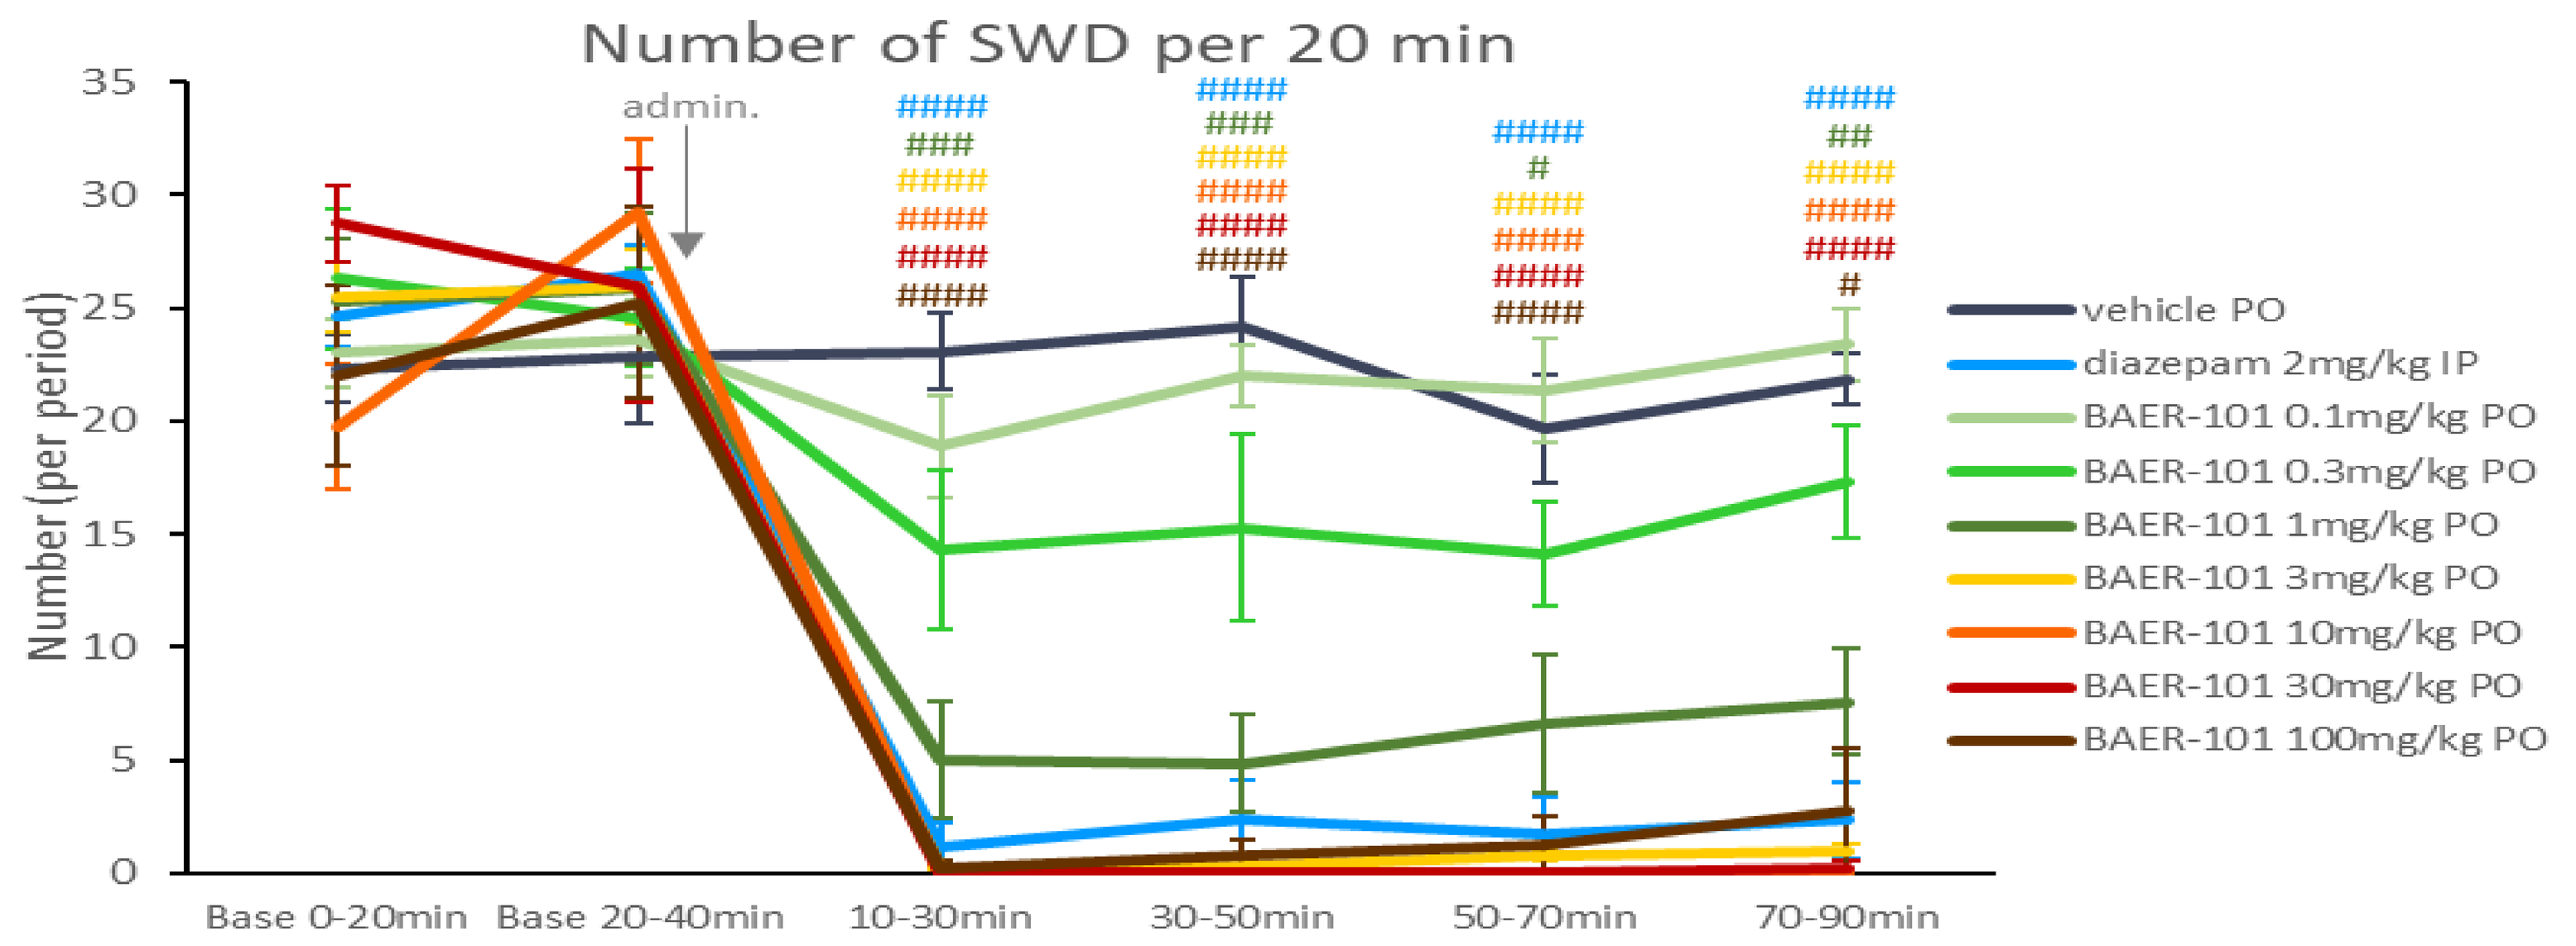
Number of SWD per 20 min
admin.
Number (per period)
0
5
10
15
20
25
30
35
Base 0-20min
Base 20-40min
10-30min
30-50min
50-70min
70-90min
vehicle PO
diazepam 2mg/kg IP
BAER-101 0.1mg/kg PO
BAER-101 0.3mg/kg PO
BAER-101 1mg/kg PO
BAER-101 3mg/kg PO
BAER-101 10mg/kg PO
BAER-101 30mg/kg PO
BAER-101 100mg/kg PO

The total time spent in SWD is a combined parameter, affected by both the number of remaining SWDs and their duration. BAER-101 significantly reduced the total time as compared to the vehicle, at all doses above 1 mg/kg (**Figure 3**). The reduction was immediately obtained, 10 to 30 min after administration, with all these doses.

Figure 3: Total time spent in SWDs per 20 min, in the vehicle condition (dark blue), diazepam at 2 mg/kg IP (light blue), and BAER-101 at 0.1 to 100 mg/kg PO (other colours). The grey arrow indicates the administration. ##, ###, ####: $p < 0.01$, 0.001 and 0.0001 as compared to vehicle

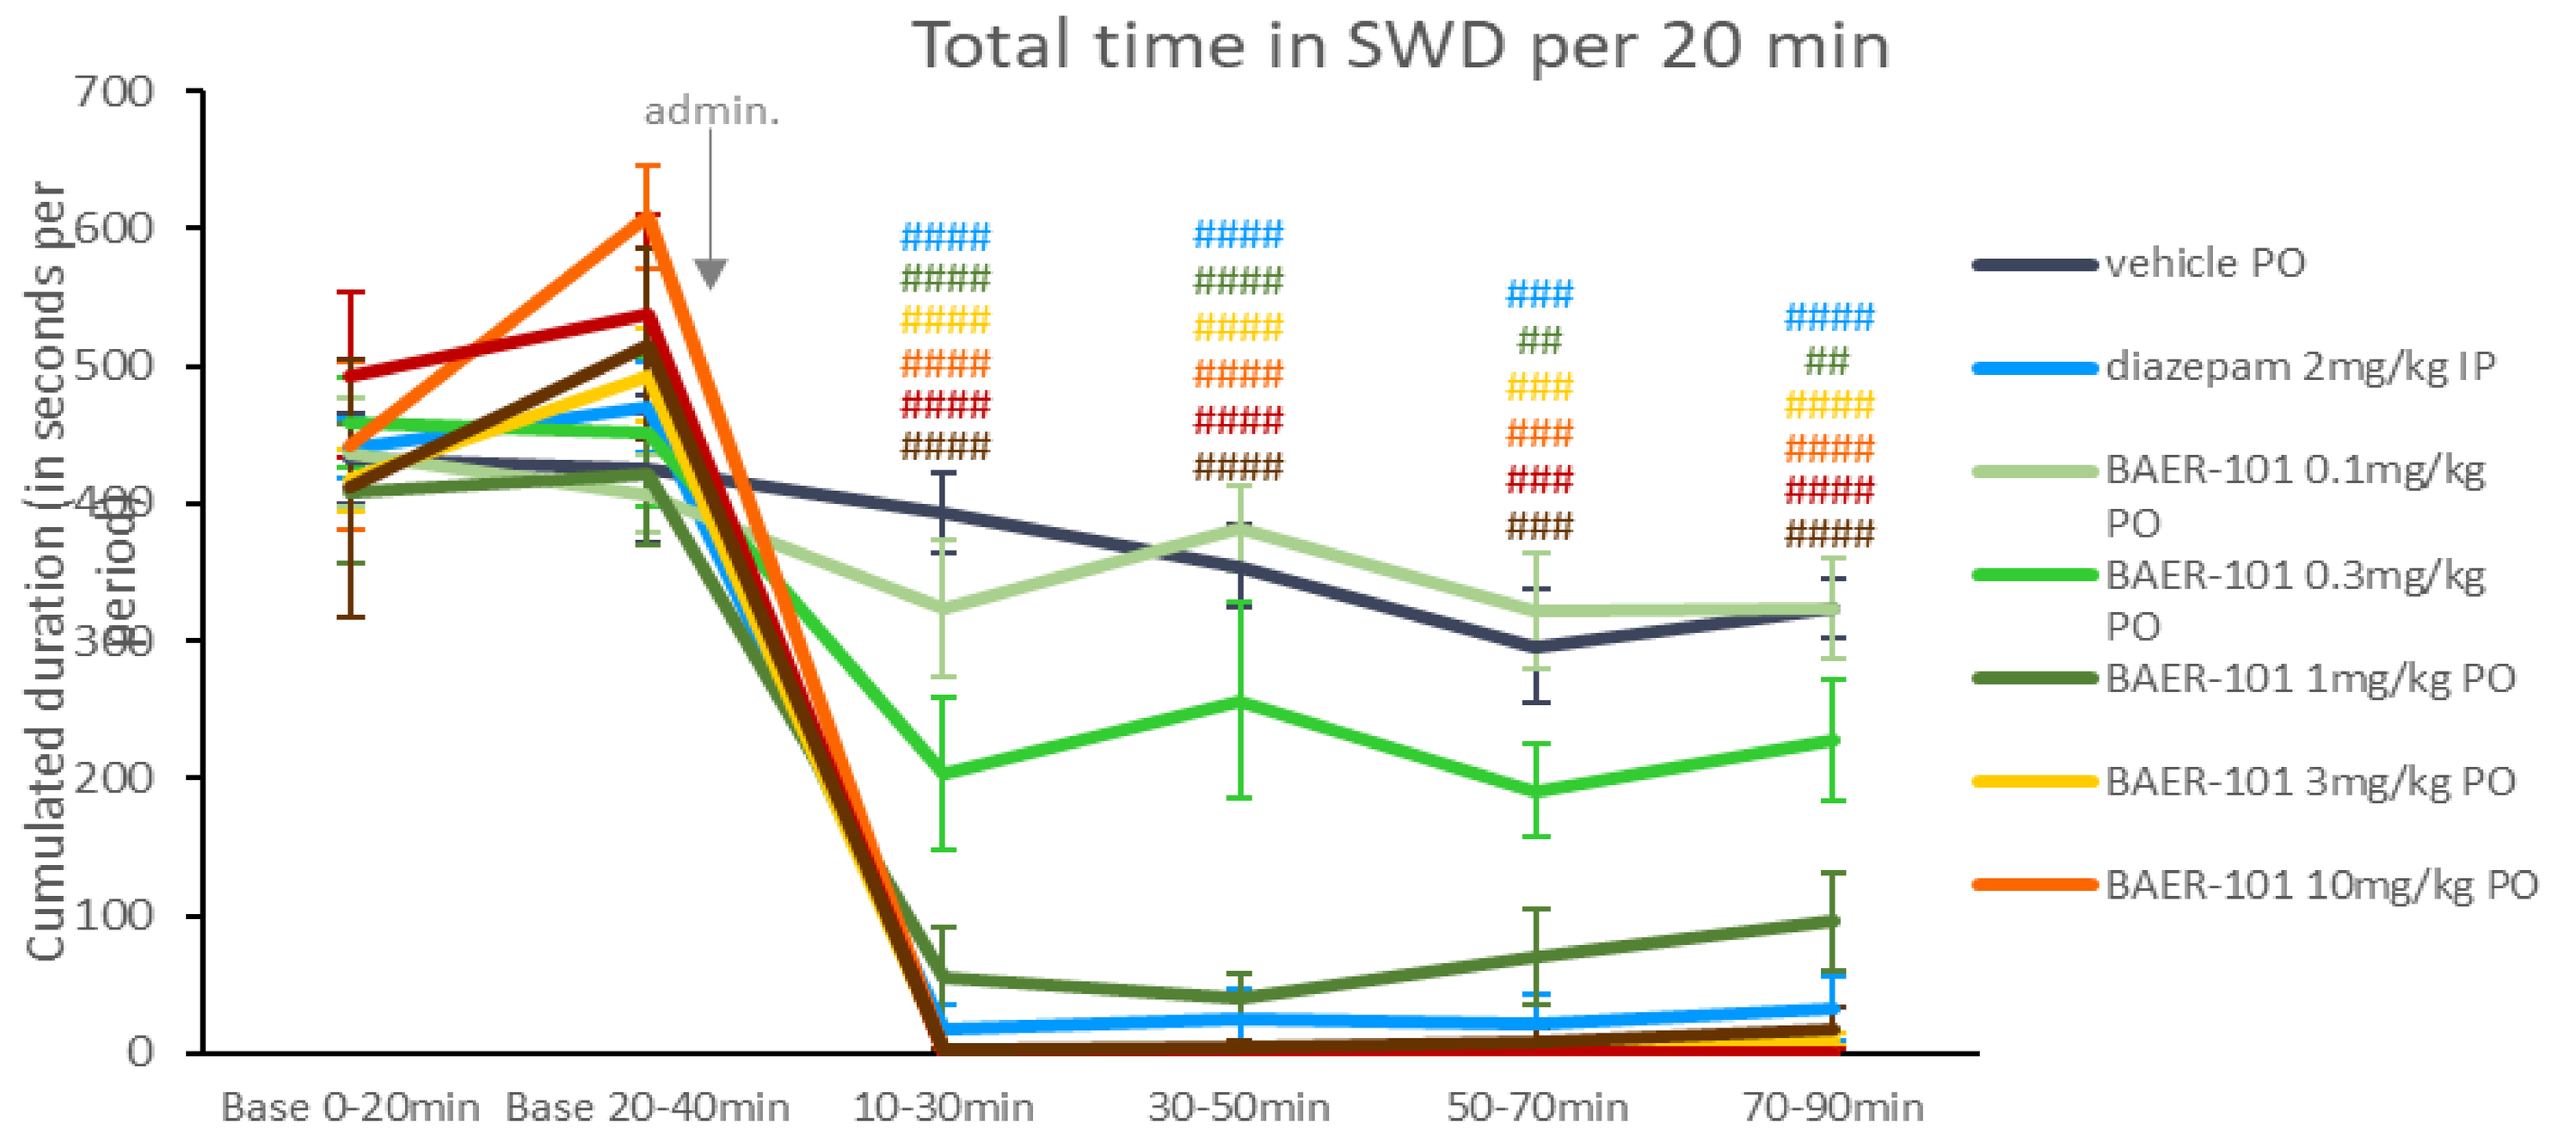
Total time in SWD per 20 min
admin.
Cumulated duration (in seconds per period)
0
100
200
300
400
500
600
700
Base 0-20min
Base 20-40min
10-30min
30-50min
50-70min
70-90min
vehicle PO
diazepam 2mg/kg IP
BAER-101 0.1mg/kg PO
BAER-101 0.3mg/kg PO
BAER-101 1mg/kg PO
BAER-101 3mg/kg PO
BAER-101 10mg/kg PO

Concentrations of BAER-101 in plasma of the rats were consistent across each EEG and blood sampling period and showed dose-dependence (**Figure 4**). The dose of orally administered BAER-101 was proportionally related to the concentrations of BAER-101 achieved in rat plasma, $p < 0.0001$. The concentration of BAER-101 in plasma was significantly associated with suppression of the number of SWD. The minimal effect dose (MED) for SWD suppression was 0.3 mg/kg which generated plasma levels in rats of 3.58 ng/mL = 10.1 nM. 10 nM exceeds the Ki for α2- and α3-containing GABAA receptors but is 23 times lower than the Ki for α5-containing GABAA receptors, demonstrating on-target engagement of BAER-101 and providing plasma levels to help guide first human dose in epileptic patients.

Figure 4: Concentrations of BAER-101 across each of the six EEG and blood sampling periods

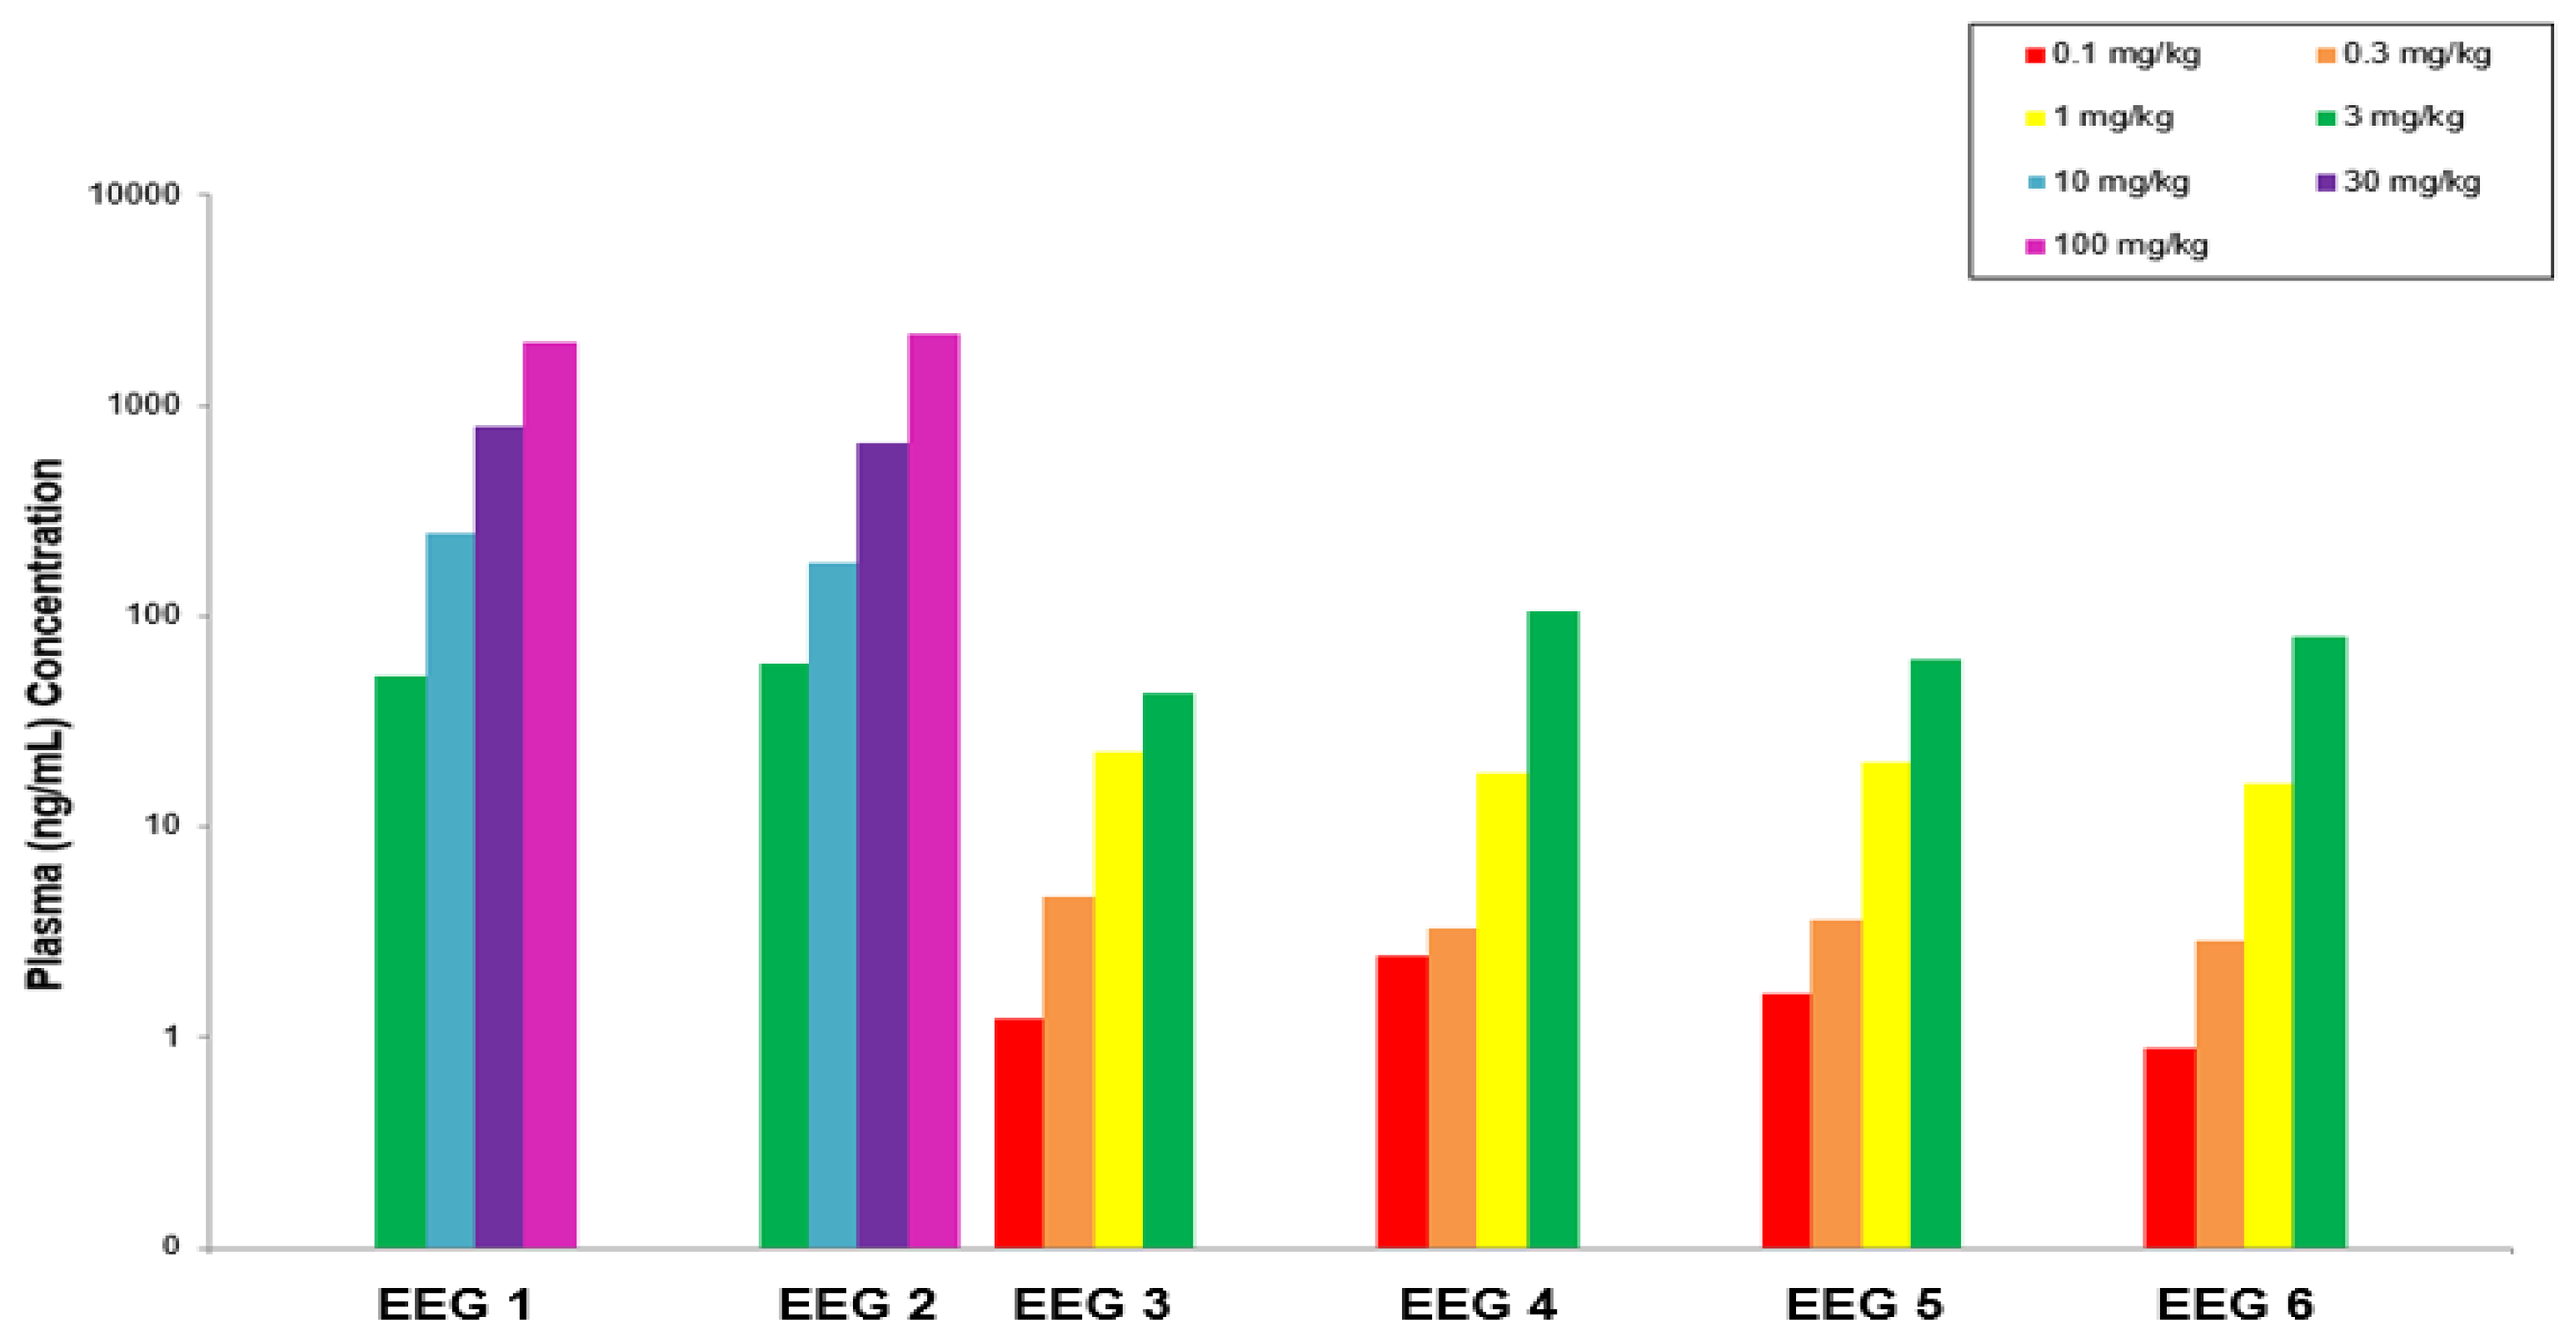
0.1 mg/kg
0.3 mg/kg
1 mg/kg
3 mg/kg
10 mg/kg
30 mg/kg
100 mg/kg
Plasma (ng/mL) Concentration
10000
1000
100
10
1
0
EEG 1
EEG 2
EEG 3
EEG 4
EEG 5
EEG 6

FIGURE 5. Concentrations of BAER-101 from rat plasma as a function of dose are shown in the left panel. The relationship of BAER-101 concentration to suppression of SWD in the GAERS model is presented on the right. The bars are means + S.D. The points are means.

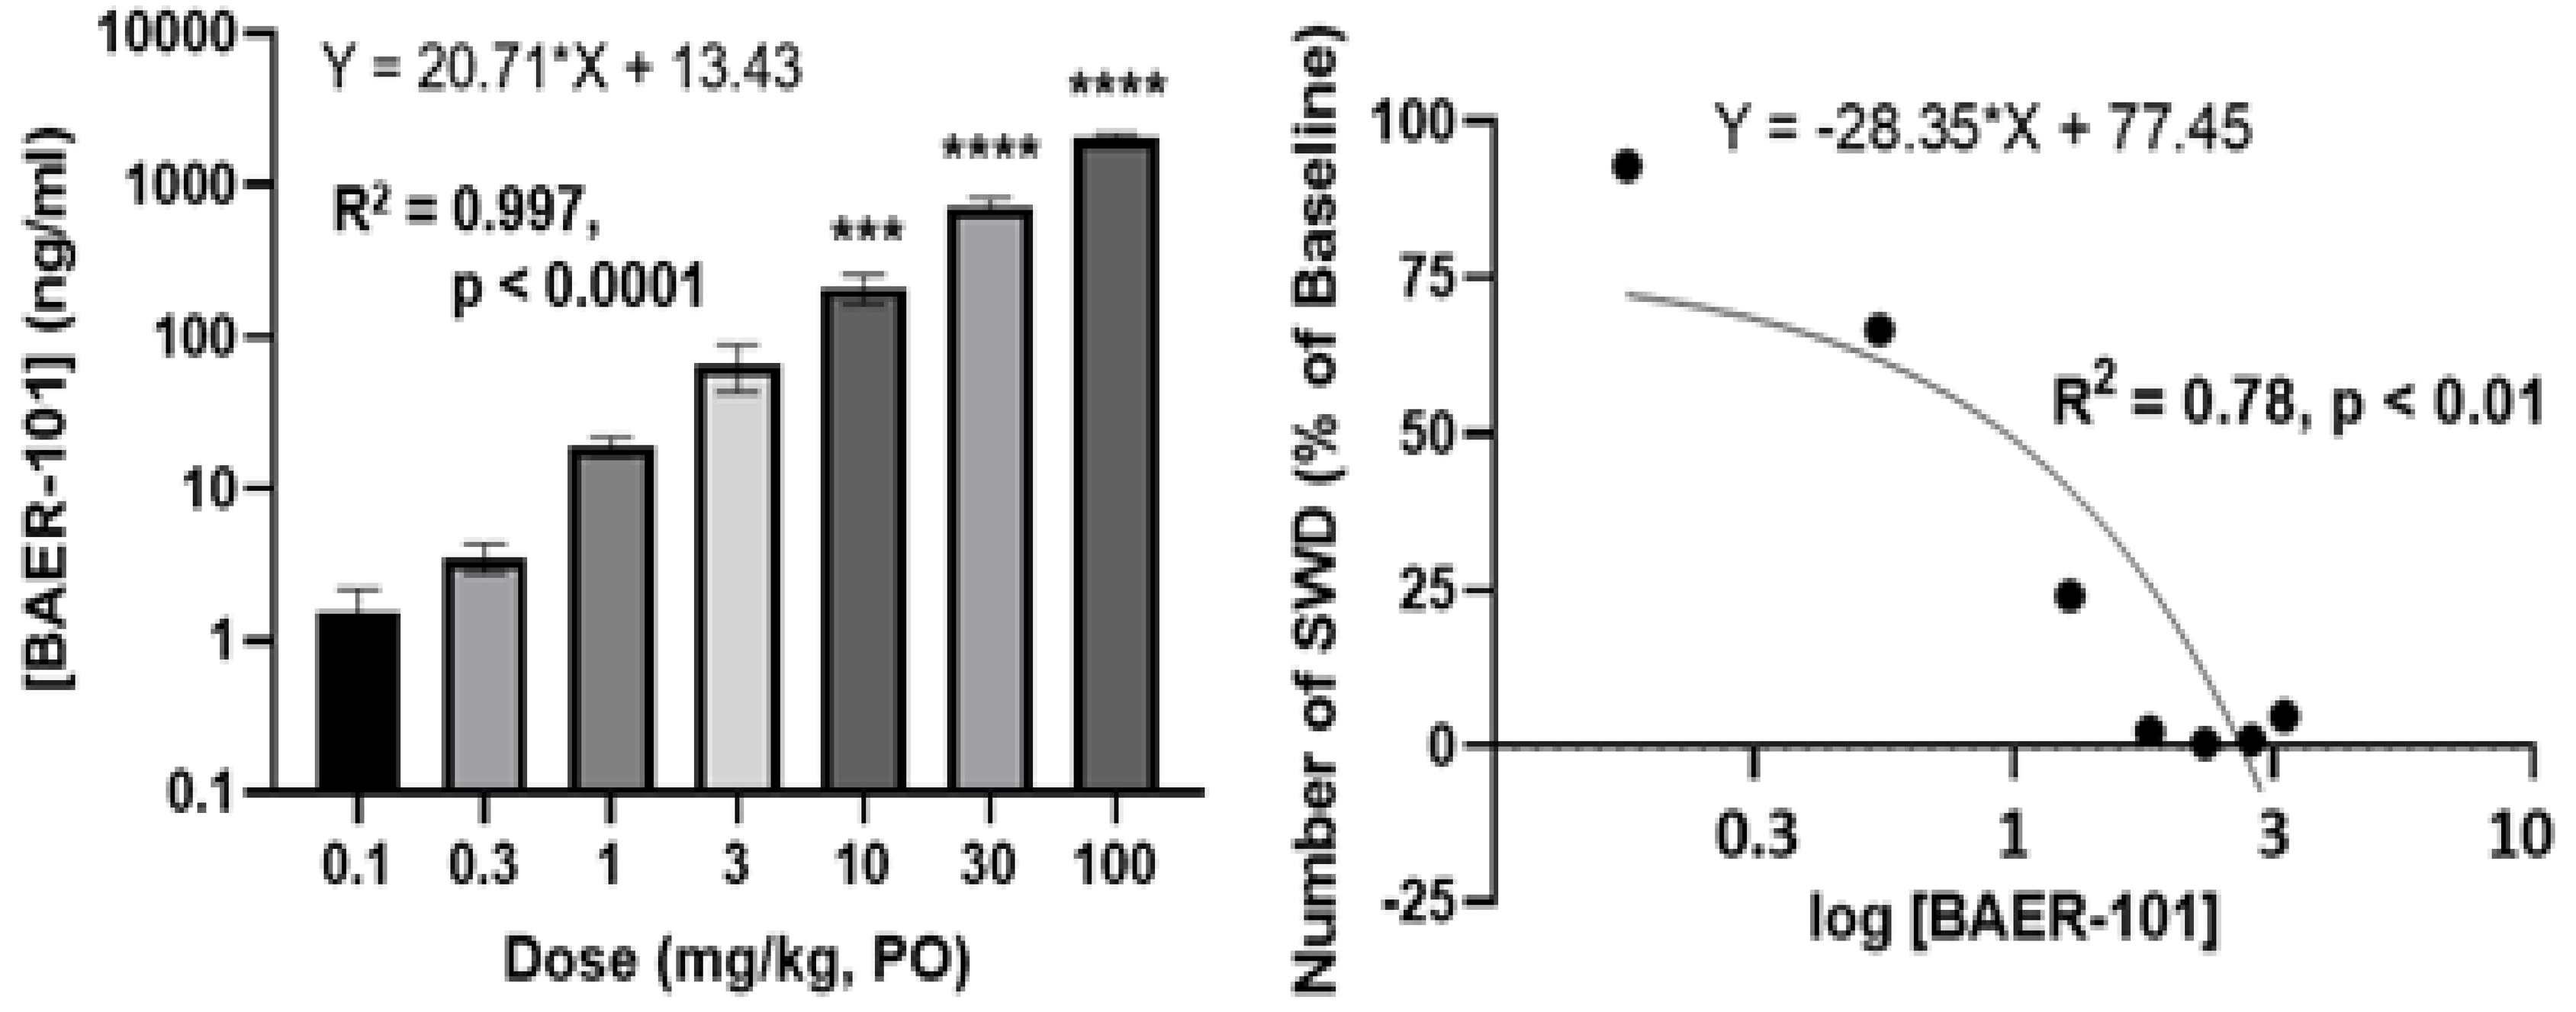
Y = 20.71*X + 13.43
R² = 0.997,
p < 0.0001
[BAER-101] (ng/ml)
10000
1000
100
10
1
0.1

0.1
0.3
1
3
10
30
100
Dose (mg/kg, PO)
Y = -28.35*X + 77.45
R² = 0.78, p < 0.01
Number of SWD (% of Baseline)
100
75
50
25
0
-25
0.3
1
3
10
log [BAER-101]

Conclusions

Based on prior patient safety and tolerability data, coupled with the high potency and full efficacy in the GAERS model, BAER-101 is considered to be a drug suitable for advancing into human clinical trials for epilepsy.

This study was funded by Avenue Therapeutics, a pharmaceutical company developing the investigational product. For further information, contact Alexandra MacLean, MD at AMacLean@avenuetx.com